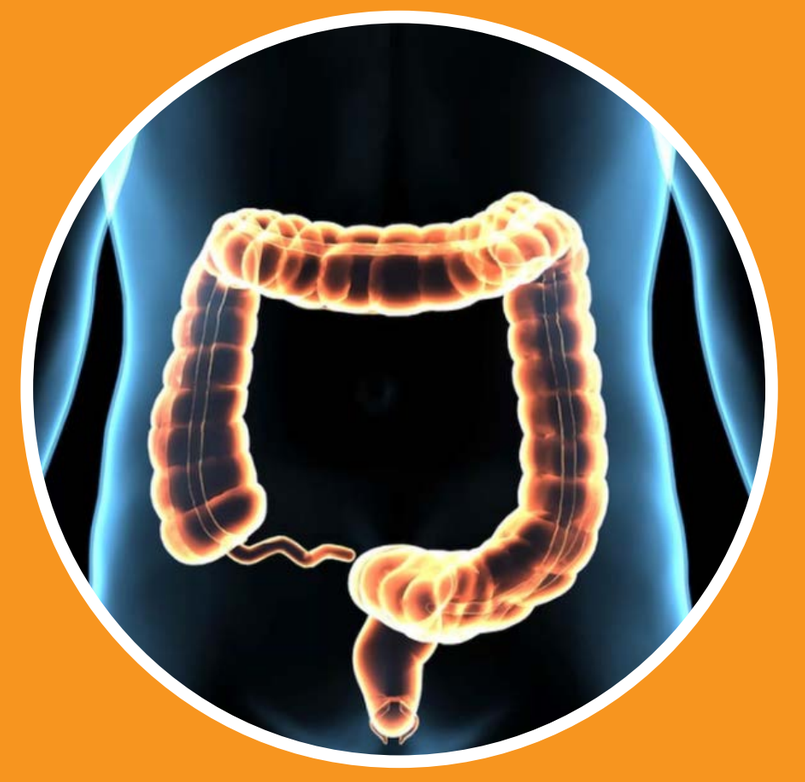

ISO-CC-005; a phase I/II study of arfolitixorin ([6R]-5,10-MTHF) in combination with 5-fluorouracil (5-FU), irinotecan and oxaliplatin ± bevacizumab in patients with metastasizing colorectal cancer

Göran Carlsson MD, PhD[1], Johan Haux MD, PhD[2], Tormod Kyrre Guren MD, PhD[3],Per Pfeiffer MD, PhD[4] Christos Papadimitrou MD, PhD[5],
Nikolaos Kentepozidis MD, PhD[6], Panagiotis Katsaounis MD[7], Dimitrios Mavroudis MD, PhD[8], Dimitrios Boumpas MD, PhD9, Karin Ganlöv, MD[10], Bengt Gustavsson MD, PhD[1]

[1]Sahlgrenska University of Gothenburg, Gothenburg, Sweden; [2]Skaraborgs Hospital, Skövde, Sweden; [3]Norwegian Radium Hospital, Oslo, Norway [4]Odense University Hospital, Odense, Denmark;
[5]Aretaieio University Hospital, Athens, Greece, [6]251 Air Force Hospital, Athens, Greece, [7]IASO General Hospital, Athens, Greece, [8]University General Hospital, Heraklion, Greece [9]University General Hospital, Athens [10] Isofol Medical AB, Göteborg, Sweden

INTRODUCTION

Chemotherapy treatment of Colorectal Cancer, often include 5-FU. 5-FU inhibits the enzyme thymidylate synthase (TS), stopping the supply of thymidine for DNA synthesis. 5-FU is always combined with a folate, which enhances the 5-FU effect. Marketed folates such as LV/L-LV are prodrugs needing enzymatic conversion (Figure 1).

Arfolitixorin (formerly called Modufolin®) is the natural, biologically active form of the folates and is expected to be efficacious in a larger proportion of patients with less inter- and intra-individual variability[1].

METHODS

ISO-CC-005 is a multi-center, phase I/II study in mCRC patients eligible for 5-FU/folate therapy alone or in combination with irinotecan or oxaliplatin ± bevacizumab.

The study investigates safety and tolerability of arfolitixorin at 4 dose levels by analysing the number and severity of AEs, SAEs and DLTs. All receives arfolitoxorin twice every two weeks during at least 4 cycles of chemotherapy. Safety is evaluated after every cycle and efficacy is evaluated after 4 cycles of chemotherapy. Gene expression, deoxyuridine levels as an indirect marker of TS inhibition and time to death is also investigated.

3-6 patients per cohort are included.

RESULTS

To date, 81 patients with mCRC have been enrolled (of which 75 have initiated treatment) in the ongoing ISO-CC-005 study. 37 SAEs, of which 9 were assessed as at least possibly related to arfolitixorin. have been reported in 20 patients. **(table 1)**, No SAE was assessed as solely related to arfolitixorin. 594 AEs of which 255 were assessed as at least possibly related to arfolitixorin have been reported in 66 patients. 1 AE (skin lesion left hand) was judged as solely related to arfolitixorin. The most frequent AEs are fatigue, nausea, neutropenia, diarrhoea, vomiting, neuropathy, UTI, mucositis, anemia and abdominal pain **(table 2)**. 334 of the AEs were grade1, 189 grade 2, 62 grade 3, 8 grade 4 and 2 grade 5. There were 2 deaths in the study, both related to progressive colorectal cancer.

After 4 cycles of treatment, 20 1st line patients have been assessed for efficacy; Early Tumour Shrinkage (ETS) and Overall response rate (ORR) according to RECIST 1.1. ETS was found in 10 patients (50%). 8 patients had partial response (PR) and 11 patients stable disease **(Figure 1)**. 39 2nd and later line patients have also been assessed for efficacy after 4 cycles of treatment. 26 patients showed clinical benefit of the therapy, i.e. partial response (PR) in 3 patients and stable disease (SD) in 23 patients. 13 patients showed progressive disease.

Table 1. Serious adverse events (SAEs)

Serious adverse events (quantity, * related to arfolitixorin)	
abdominal pain (2)	Infection*
anal infection	obstipation (2)
ascites (2)	pain, rectal
cholangitis	Pancolitis*
Dehydration* (2)	pleural effusion
elevated levels of creatinine*	pneumothorax
epileptic seizure	progression of colorectal cancer* (2)
febrile neutropenia*	renal failure* (2)
fever (3)	retinal detachment
Hyponatremia*	stomach pain
Ileitis*	supraventricular tachycardia
ileus (4)	thrombosis vena mesenterica superior
increased CRP	urinary retention

Table 2. Most frequent adverse events (AEs)

Most frequent Adverse Events (quantity)	
Fatigue	46
Nausea	46
Neutropenia	37
Diarrhea	33
Vomiting	32
Neuropathy	30
UTI	15
Mucositis	15
Anemia	14
Abdominal pain	12

Figure 1. ETS and ORR in first line patients in the ISO-CC-005 study after 4 cycles (8 weeks) of treatment

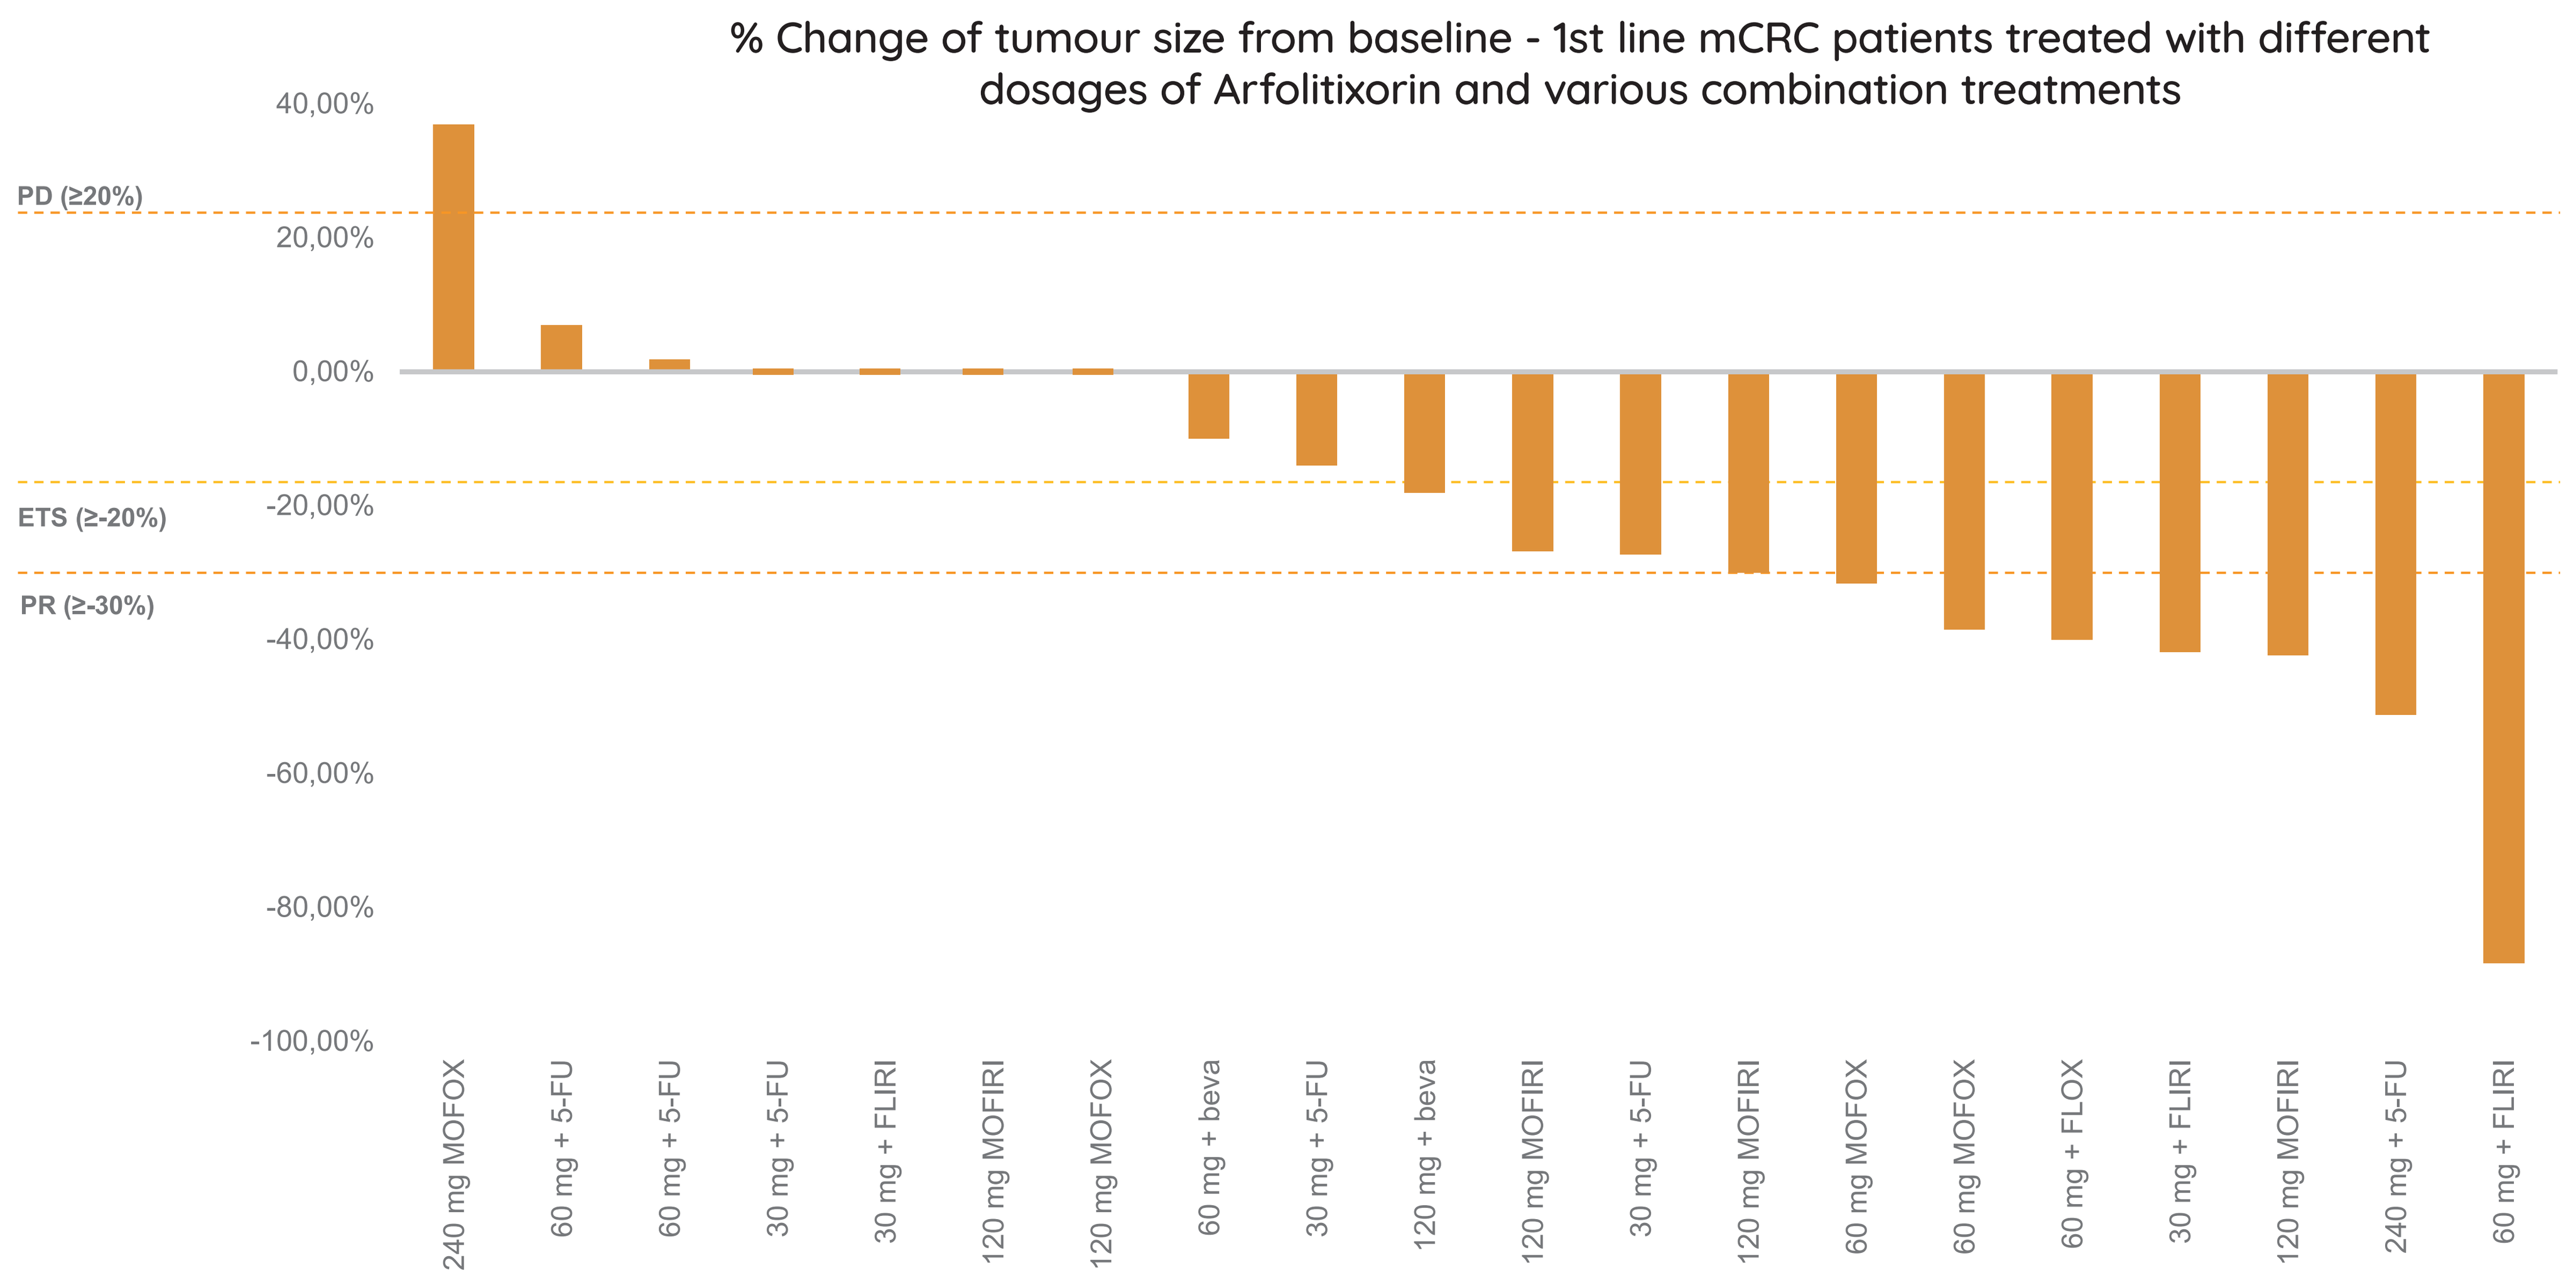
% Change of tumour size from baseline - 1st line mCRC patients treated with different dosages of Arfolitixorin and various combination treatments
40,00%
20,00%
0,00%
-20,00%
-40,00%
-60,00%
-80,00%
-100,00%
PD (≥20%)
ETS (≥-20%)
PR (≥-30%)
240 mg MOFOX
60 mg + 5-FU
60 mg + 5-FU
30 mg + 5-FU
30 mg + FLIRI
120 mg MOFIRI
120 mg MOFOX
60 mg + beva
30 mg + 5-FU
120 mg + beva
120 mg MOFIRI
30 mg + 5-FU
120 mg MOFIRI
60 mg MOFOX
60 mg MOFOX
60 mg + FLOX
30 mg + FLIRI
120 mg MOFIRI
240 mg + 5-FU
60 mg + FLIRI

Among 20 first line patients in ISO-CC-005:

Early Tumour Shrinkage:

- 10 patients with ETS (≥-20%)

Objective Response Rate*:

- 8 patients with partial response, PR (≥-30%)
- 11 patients with stable disease, SD (+20% to -30%)
- 1 patient with progressed disease, PD (≥20%)

* According to RECIST 1.1.

CONCLUSIONS

The lack of need for metabolic activation makes arfolitixorin a better candidate than LV/L-LV for improved outcome of 5-FU-based chemotherapy regimens in mCRC.

The ISO-CC-005 study evaluates arfolitixorinin combination with 5-FU, irinotecan, oxaliplatin ± bevacizumab in mCRC patients in 4 countries in Europe.

The results, so far, in patients treated with different doses of arfolitixorin and in different treatment combinations seems promising for both safety and efficacy.

A multicentre study comparing efficacy of arfolitixorin and LV in a standard chemotherapy/target drug combination in 1st line metastatic CRC patients is planned.

More information about the study is available at www.clinicaltrials.gov

Figure 1. Mechanism of action of arfolitixorin

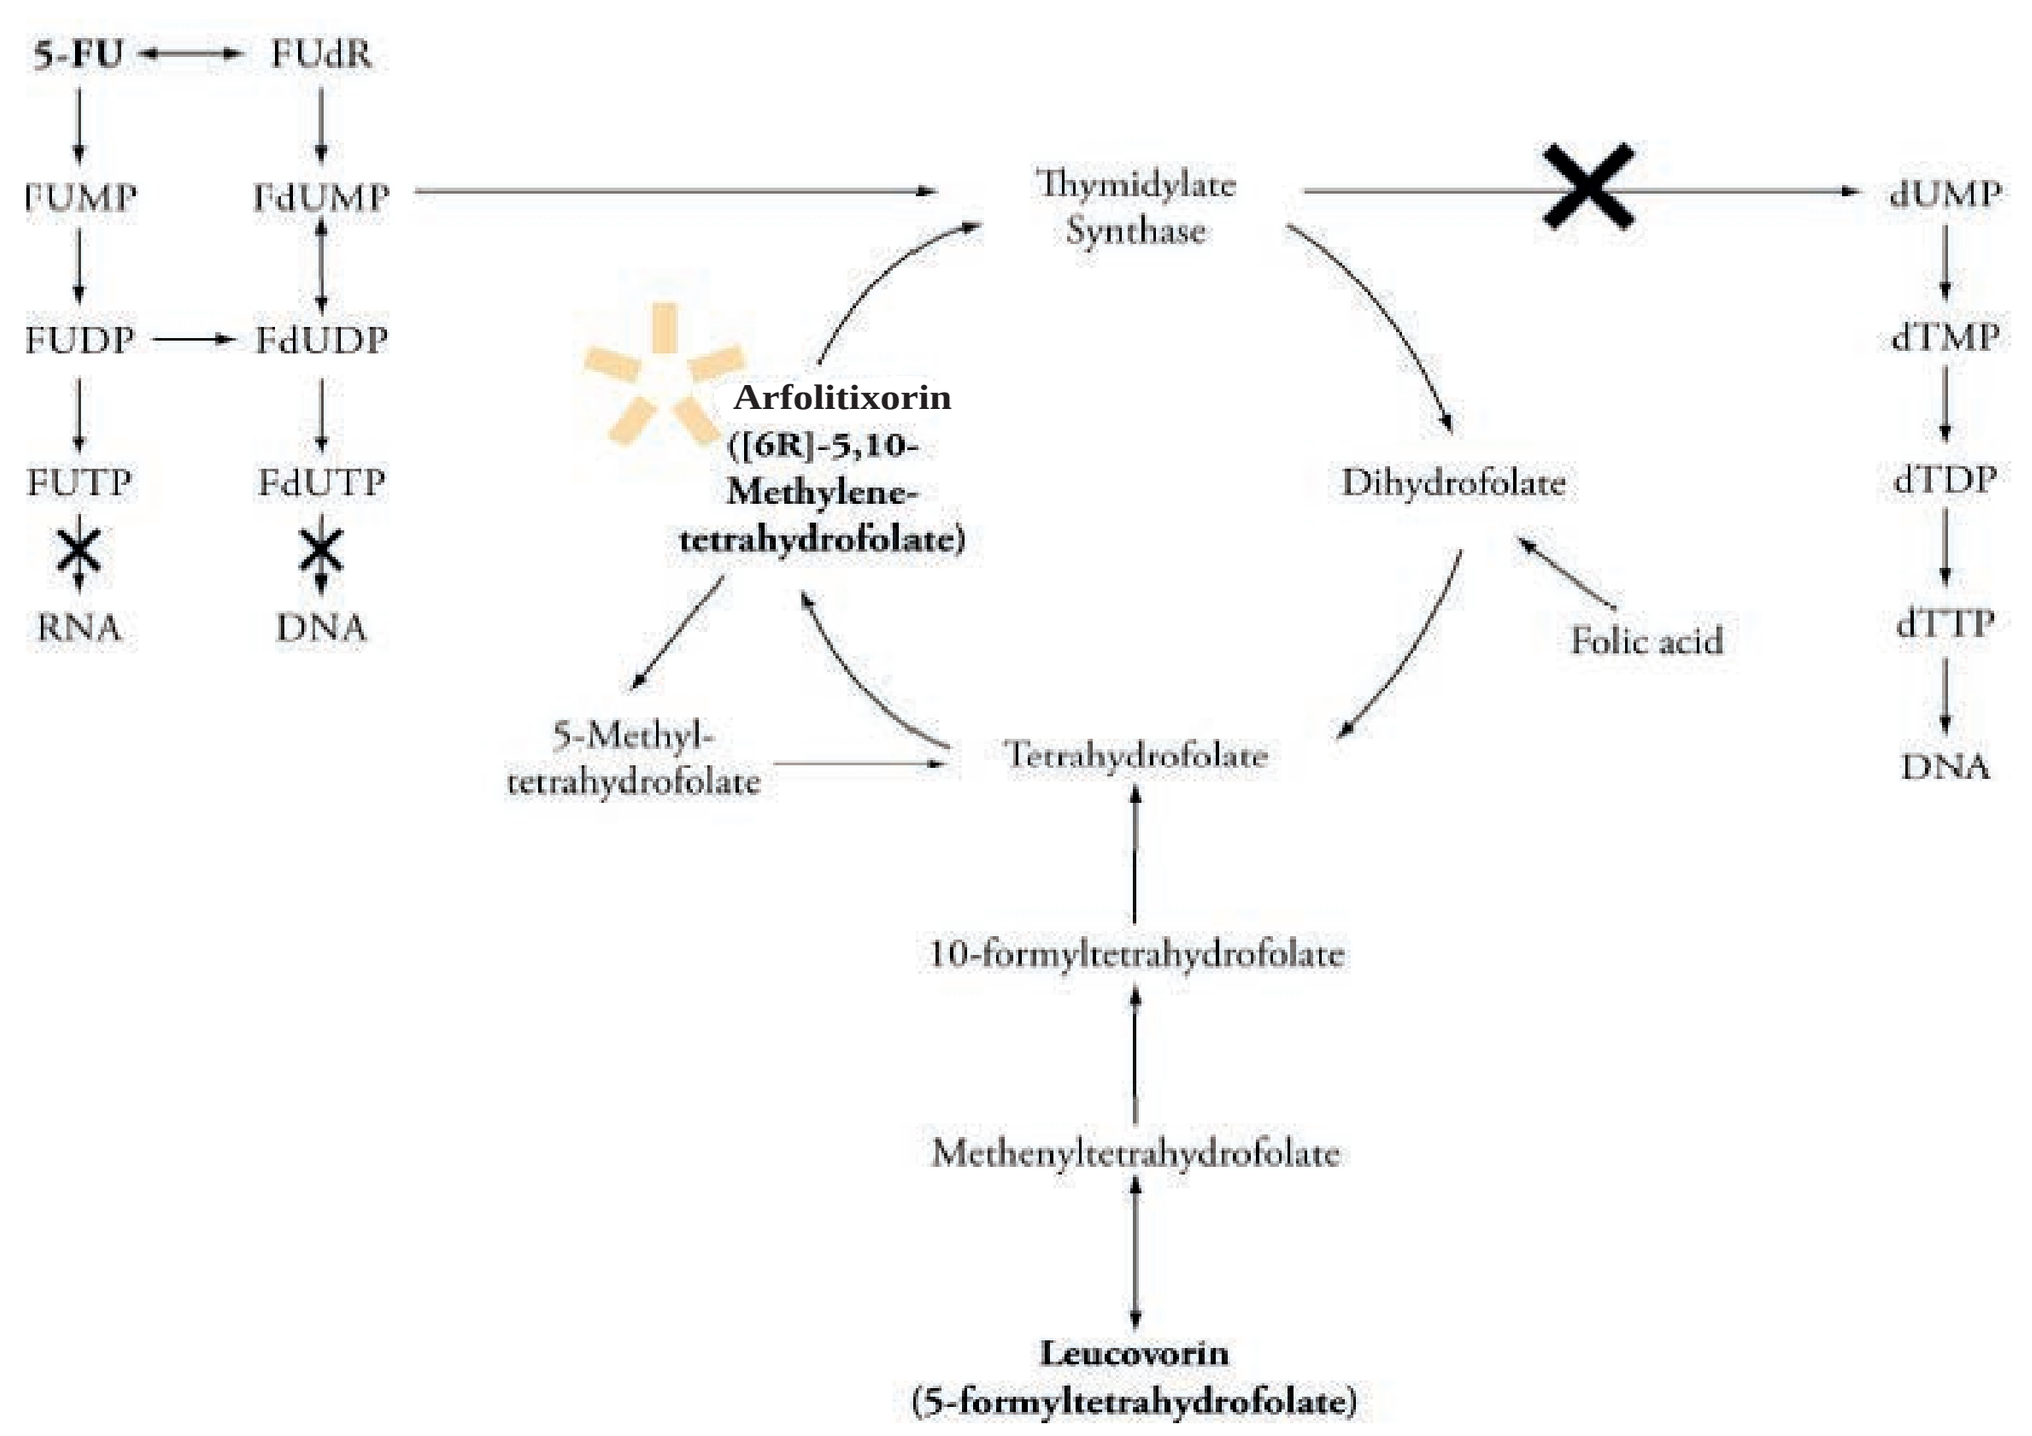
5-FU
FUdR
FUMP
FdUMP
FUDP
FdUDP
FUTP
FdUTP
RNA
DNA
Thymidylate Synthase
dUMP
dTMP
dTDP
dTTP
DNA
Arfolitixorin ([6R]-5,10-Methylene-tetrahydrofolate)
Dihydrofolate
Folic acid
5-Methyl-tetrahydrofolate
Tetrahydrofolate
10-formyltetrahydrofolate
Methenyltetrahydrofolate
Leucovorin (5-formyltetrahydrofolate)

References

1. Danenberg PV et al. Folates as adjuvants to anticancer agents: Chemical rationale and mechanism of action. Crit Rev Oncol Hematol 2016; 106:118-131

Poster presented at ESMO 2018 Congress. Poster no.569P
Contact person Dr Göran Carlsson, e-mail address goran.u.carlsson@vgregion.se