Abstract 205: The Importance of Treatment Handling and Compliance on Overall Response Rate in a Phase III Study of Metastatic Colorectal Cancer (Post-hoc Per Protocol Analyses of the AGENT Trial)

Aldina Pivodic PhD (aldina.pivodic@apnc.se), Göran Carlsson MD PhD, Åke Hjalmarson MD PhD

Background

In the AGENT trial (NCT03750786) the primary endpoint of superiority for overall response rate (ORR) was not reached (48% for arfolitixorin vs 49% for leucovorin) in the ITT population (Tabernero et al 2023). The aim of the current study was to evaluate the compliance in handling the investigational medicinal product and its impact on outcomes in patients with metastatic colorectal cancer participating in the AGENT trial.

Methods

In a randomized, multicenter, multinational, phase III study, patients were randomized to receive either arfolitixorin (n=245) or leucovorin (n=245) with 5-fluorouracil (5-FU), oxaliplatin and bevacizumab as a first-line treatment. Logistic regression was applied for overall response rate (ORR) and Cox regression for all-cause death, and all-cause death or progression, adjusted for randomization strata.

Results

Missing or incorrect IMP handling was reported for 28 (11%) patients in the arfolitixorin and 23 (9%) in the leucovorin arm, <80% compliance for duration of bolus 5-FU 2-4min for 101 (42%) and 97 (41%), respectively. Considering the time between the bolus 5-FU and arfolitixorin, non-compliance, defined by <80% doses given within 25-35min PP window, was reported for 92 (38%) in arfolitixorin patients. Adherence to the timing between the first and second arfolitixorin dose (30-60min) was high. In total, 91 (37%) patients in the arfolitixorin and 134 (55%) in the leucovorin arm fulfilled PP definitions (cPP population).
The bolus 5-FU dose was equally likely to be reduced in both the arfolitixorin and leucovorin arms across regions (ranging from 22% to 39%), except for Japan where 55% of patients in the arfolitixorin arm had their bolus 5-FU dose reduced, compared to 28% in the leucovorin arm, despite similar type, grade and frequency of toxicity.
In the cPP population, the ORR was achieved by 54 (59%) in the arfolitixorin and 69 (51%) in the leucovorin arm, adjusted odds ratio (aOR) 1.40 (95% CI 0.81-2.45), p=0.23. Excluding Japan, the ORR was 41 (62%) in the arfolitixorin and 49 (46%) in the leucovorin arm, aOR 2.11 (95% CI 1.09-4.06), p=0.026. Corresponding aOR for Europe was 1.50 (95% CI 0.71-3.15), p=0.29, and for North America aOR 7.95 (95% CI 1.45-43.62), p=0.017. In Japan, the ORR had better outcome in the leucovorin arm (71.4%) compared to the arfolitixorin arm (52.0%), aOR 0.42 (95% CI 0.13-1.31), p=0.14. The relationship was numerically opposite for patients without bolus 5-FU dose reduced.
All-cause death during the complete follow-up was reported for 37 (41%) in the arfolitixorin and 61 (46%) in the leucovorin arm.

UNIVERSITY OF GOTHENBURG

This study was funded by Isofol Medical.

CONCLUSION: In the AGENT trial, 46% of the included patients (37% in the arfolitixorin and 55% in the leucovorin arm) met **per protocol compliance for treatment handling**. In this cPP population, there was a non-significant, but clinically relevant, increase in the overall response rate in the arfolitixorin compared to the leucovorin arm. Notably, **in all regions excluding Japan**, the overall response rate was higher for arfolitixorin, with statistical significance. Adherence to the treatment protocol is crucial for the outcomes of patients with metastatic colorectal cancer.

cPP by Region (ITT)

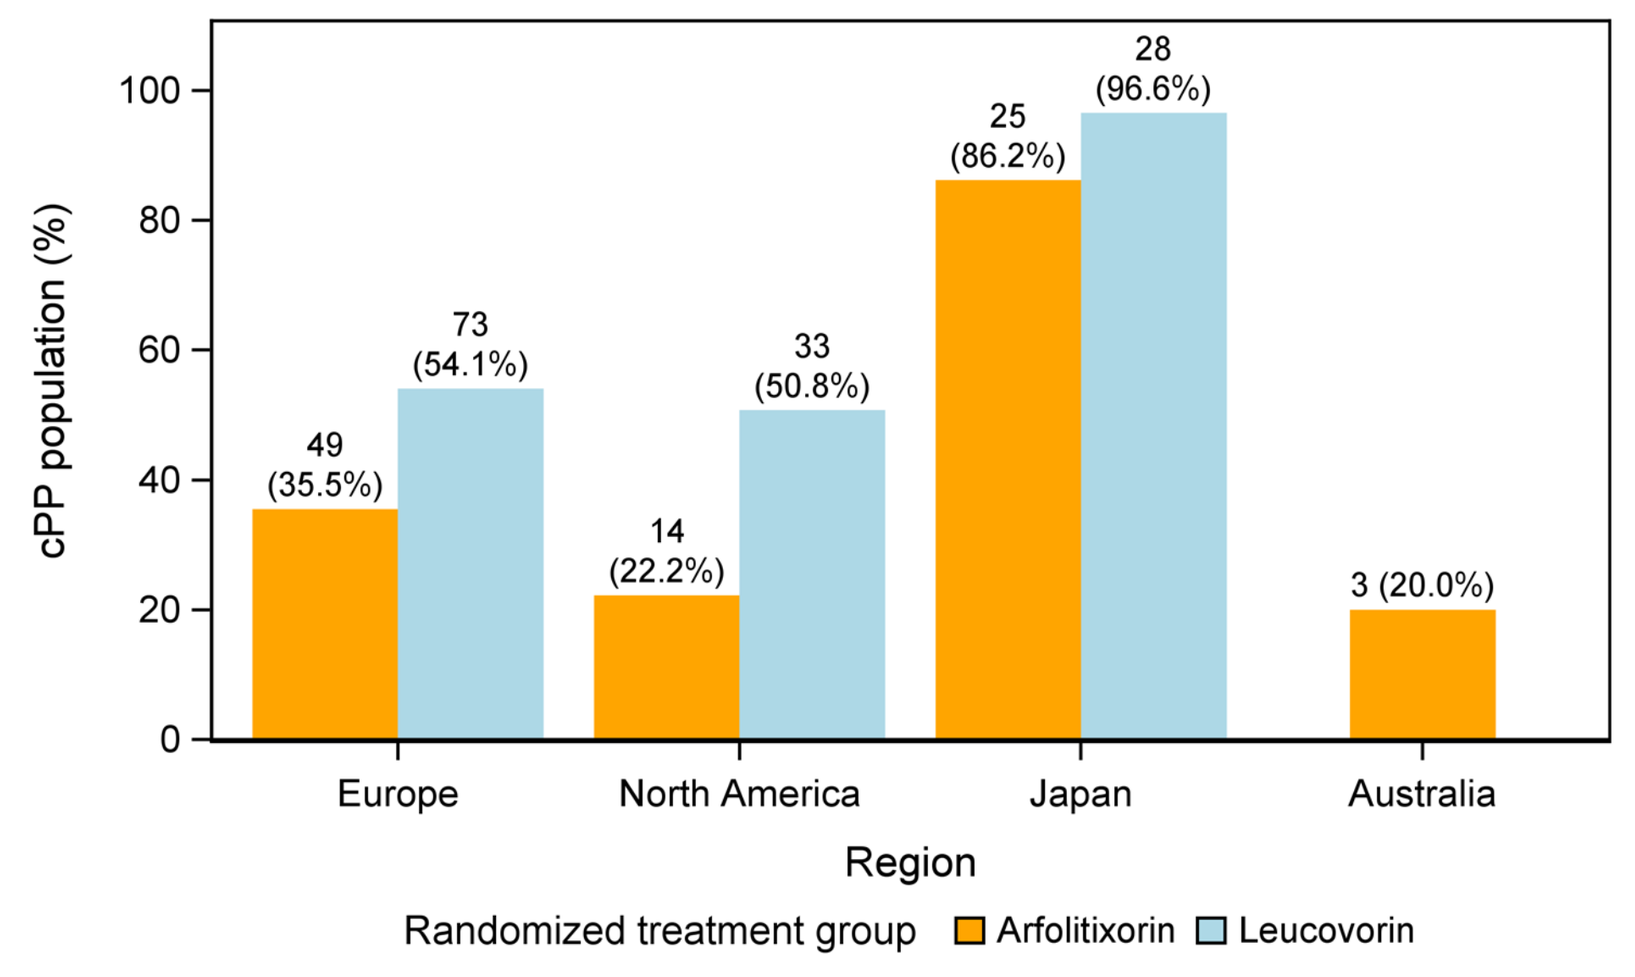
cPP population (%)
49 (35.5%)
73 (54.1%)
14 (22.2%)
33 (50.8%)
25 (86.2%)
28 (96.6%)
3 (20.0%)
Europe
North America
Japan
Australia
Region
Randomized treatment group Arfolitixorin Leucovorin

Dose Reduction (cPP)

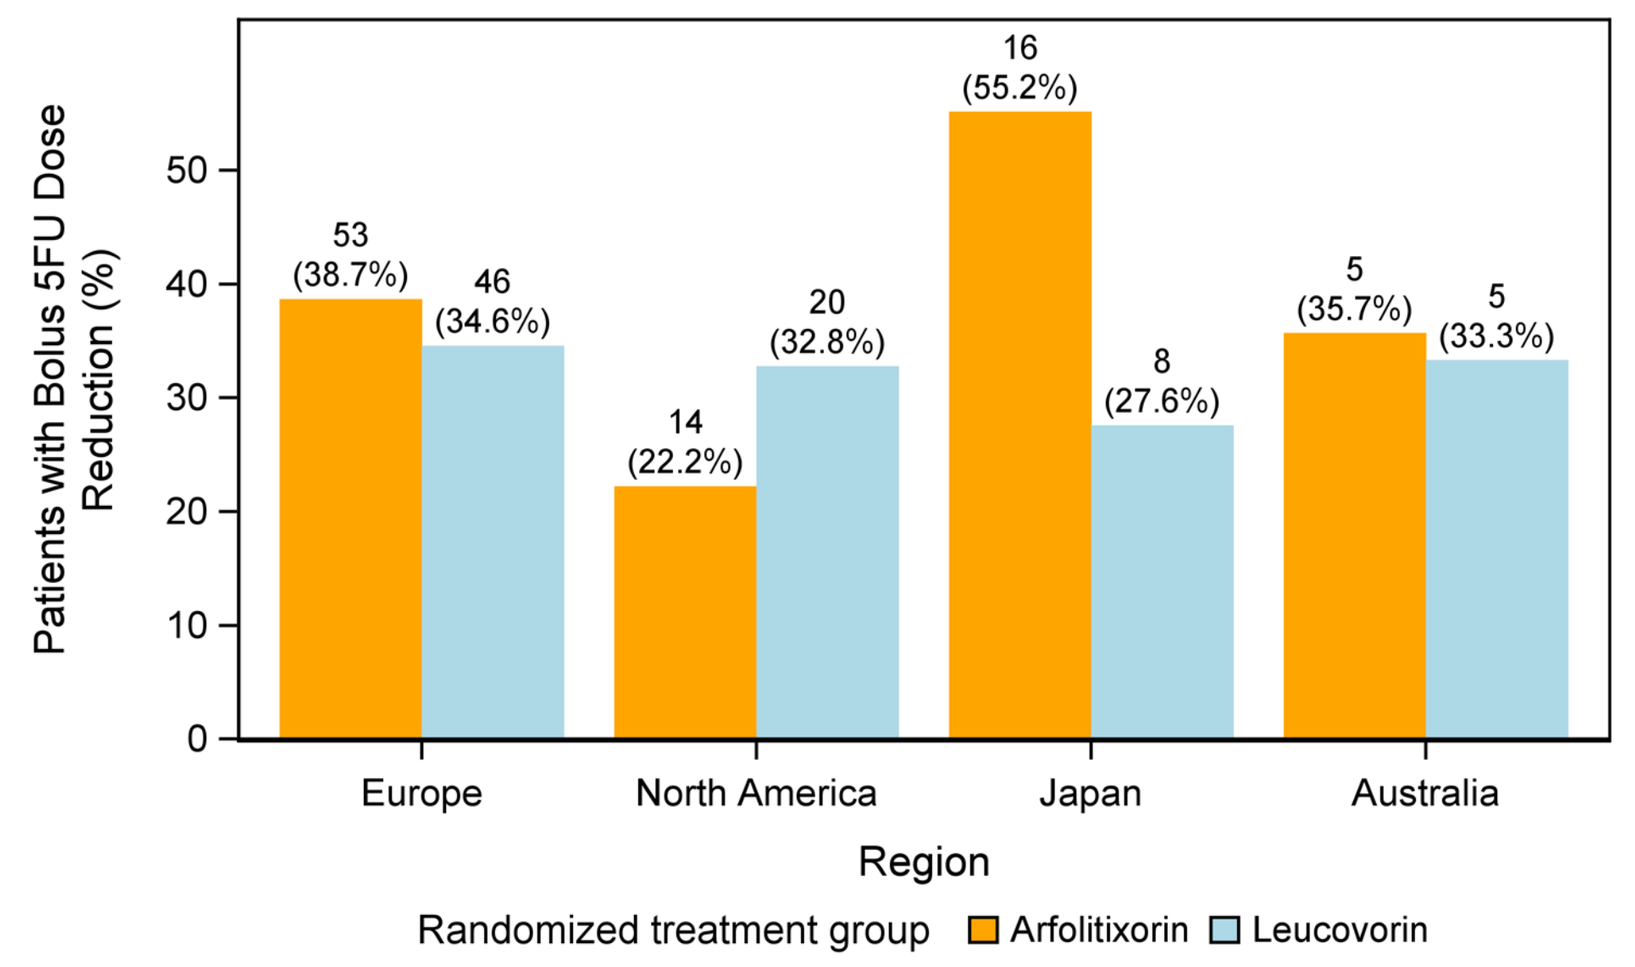
Patients with Bolus 5FU Dose Reduction (%)
53 (38.7%)
46 (34.6%)
14 (22.2%)
20 (32.8%)
16 (55.2%)
8 (27.6%)
5 (35.7%)
5 (33.3%)
Europe
North America
Japan
Australia
Region
Randomized treatment group Arfolitixorin Leucovorin

Overall Response Rate by Region (cPP)

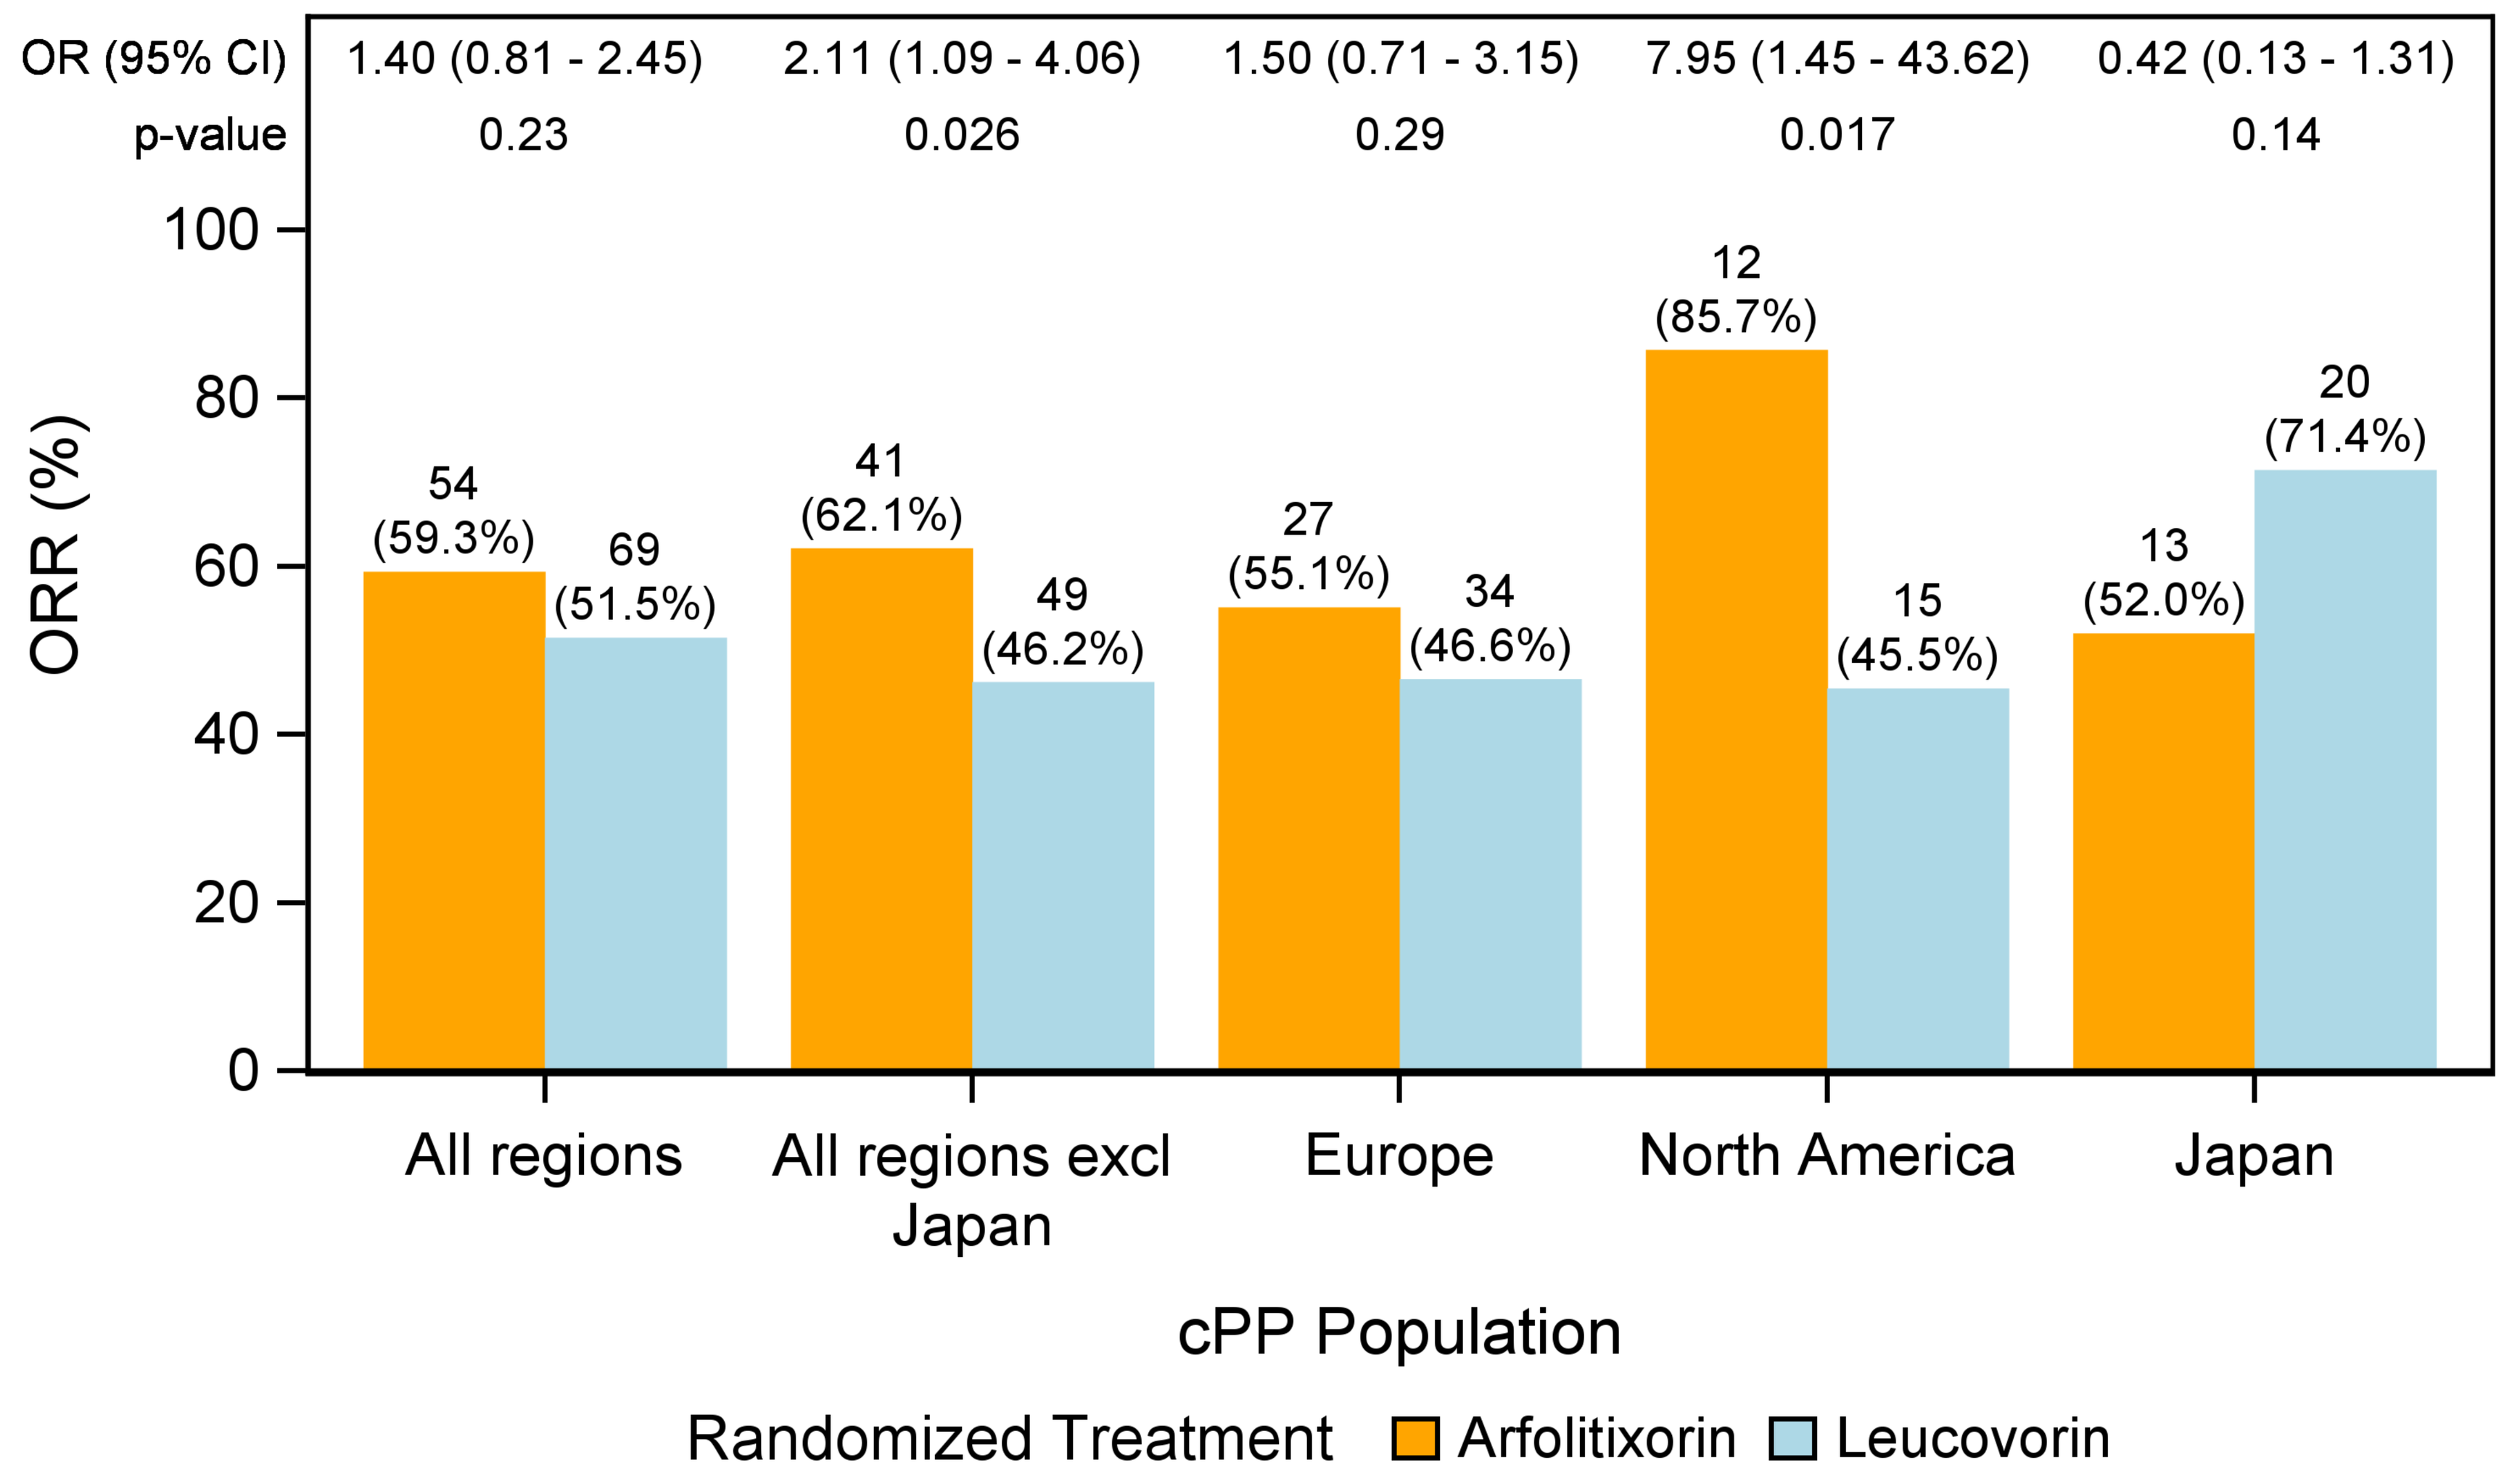
OR (95% CI)
p-value
1.40 (0.81 - 2.45)
0.23
2.11 (1.09 - 4.06)
0.026
1.50 (0.71 - 3.15)
0.29
7.95 (1.45 - 43.62)
0.017
0.42 (0.13 - 1.31)
0.14
ORR (%)
54 (59.3%)
69 (51.5%)
41 (62.1%)
49 (46.2%)
27 (55.1%)
34 (46.6%)
12 (85.7%)
15 (45.5%)
13 (52.0%)
20 (71.4%)
All regions
All regions excl Japan
Europe
North America
Japan
cPP Population
Randomized Treatment Arfolitixorin Leucovorin

ORR (cPP – No Dose Reduction)

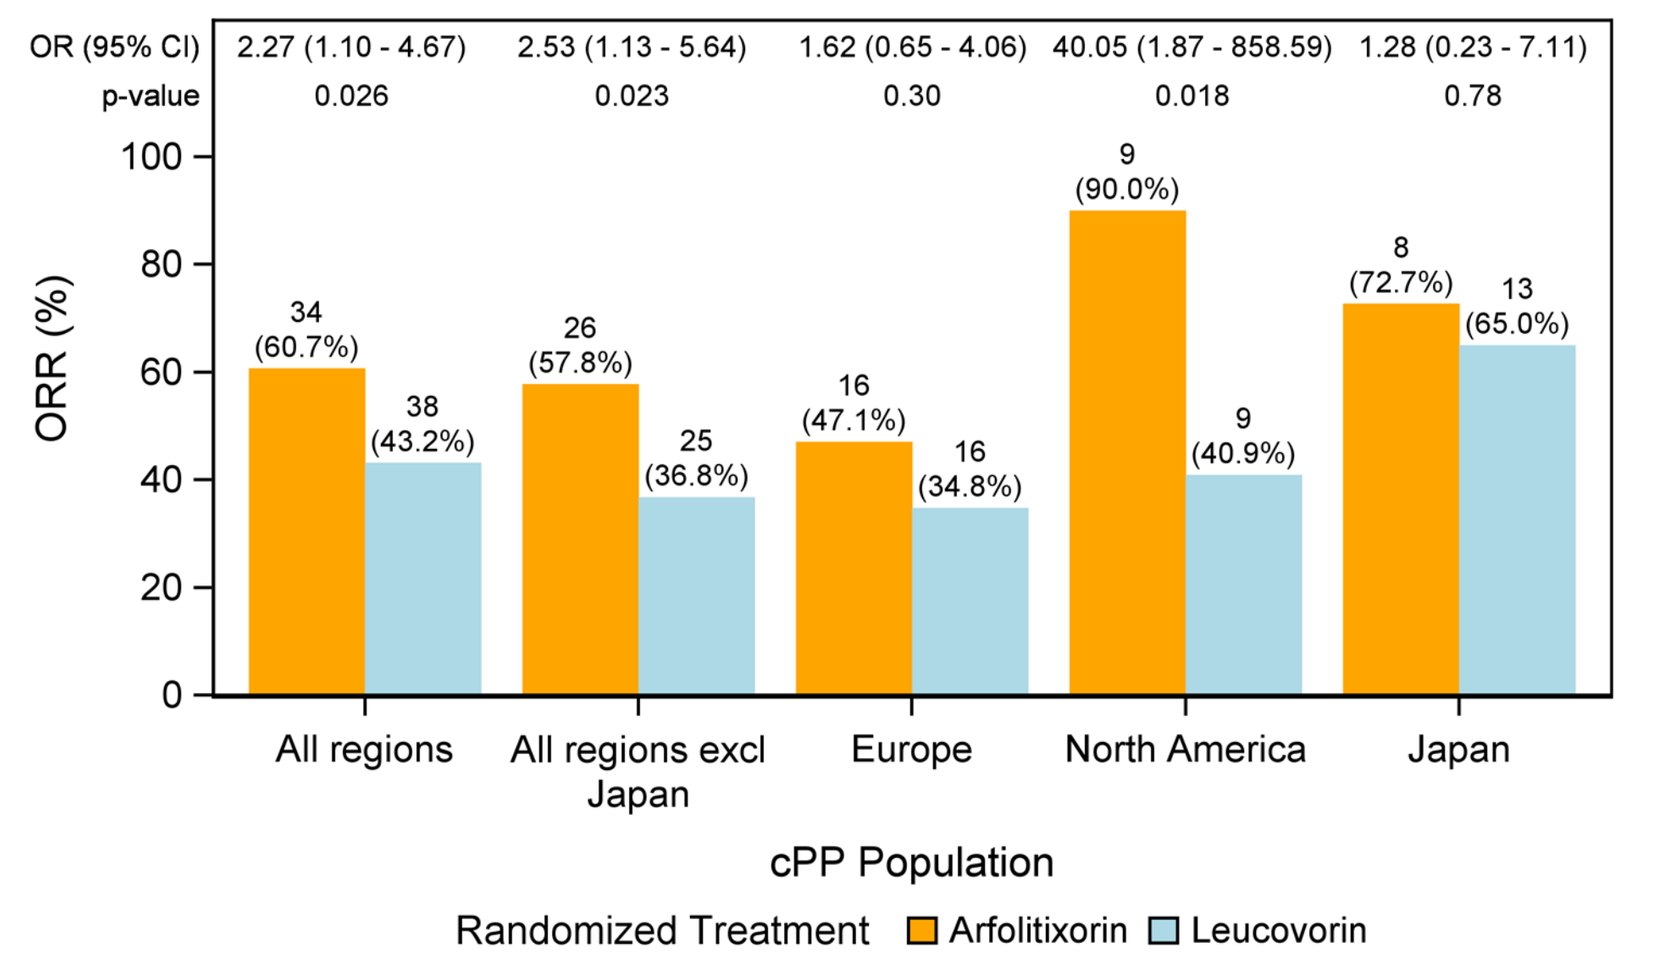
OR (95% CI)
p-value
2.27 (1.10 - 4.67)
0.026
2.53 (1.13 - 5.64)
0.023
1.62 (0.65 - 4.06)
0.30
40.05 (1.87 - 858.59)
0.018
1.28 (0.23 - 7.11)
0.78
ORR (%)
34 (60.7%)
38 (43.2%)
26 (57.8%)
25 (36.8%)
16 (47.1%)
16 (34.8%)
9 (90.0%)
9 (40.9%)
8 (72.7%)
13 (65.0%)
All regions
All regions excl Japan
Europe
North America
Japan
cPP Population
Randomized Treatment Arfolitixorin Leucovorin

ORR (cPP – Dose Reduction)

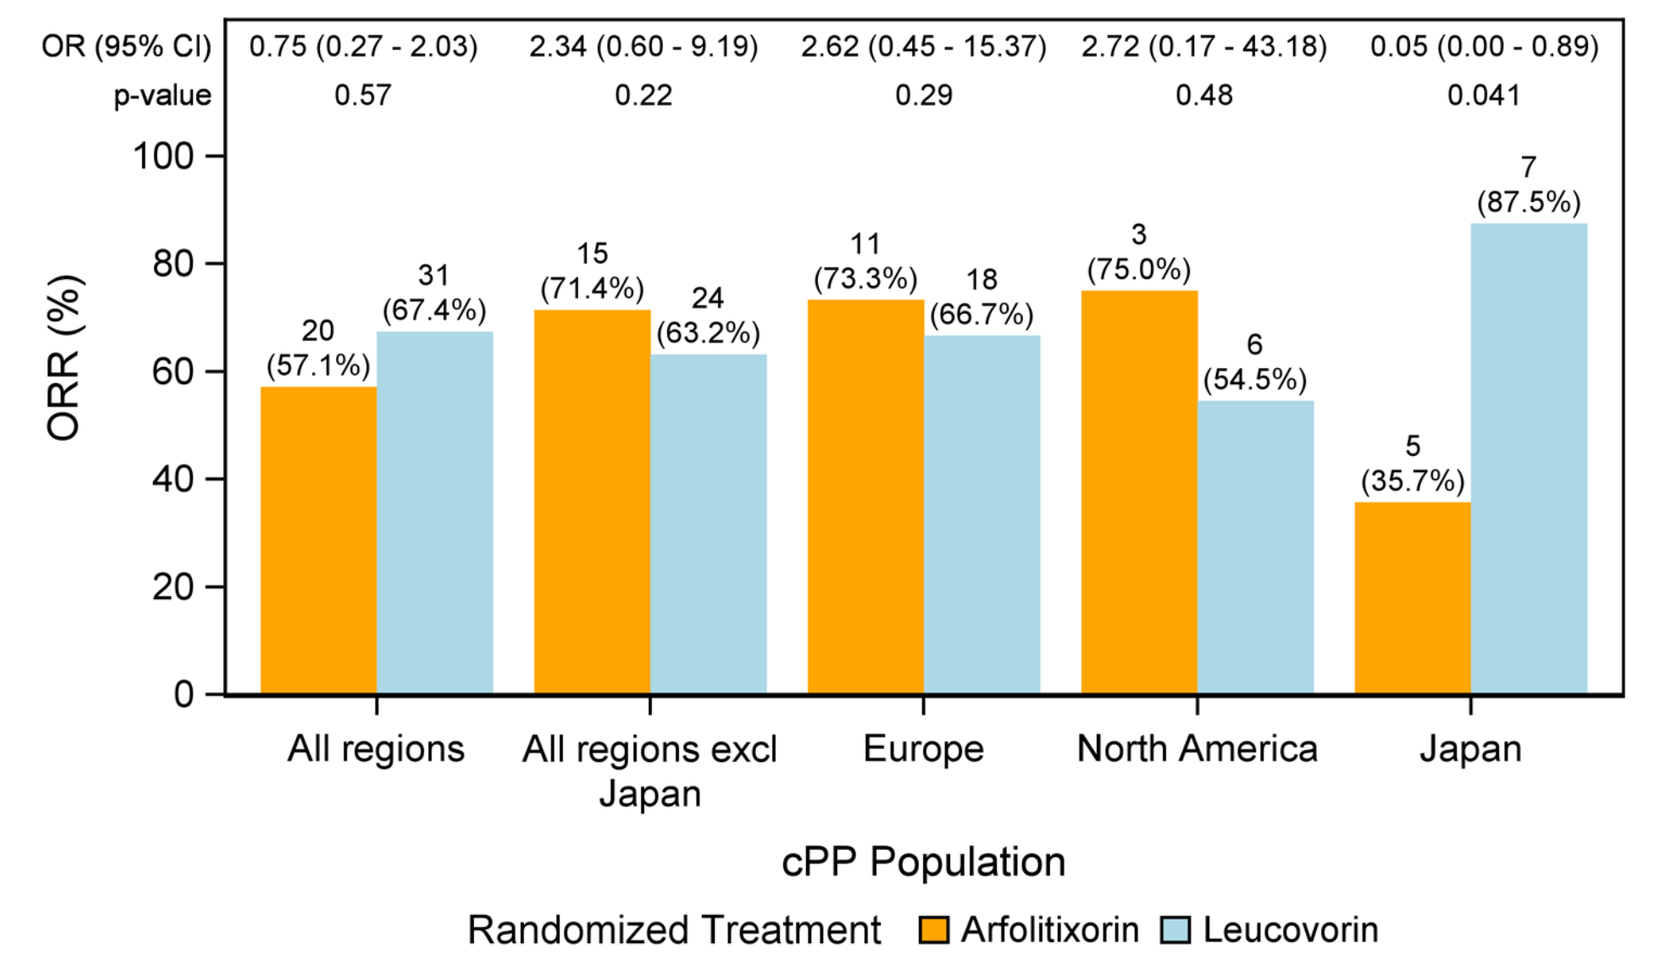
OR (95% CI)
p-value
0.75 (0.27 - 2.03)
0.57
2.34 (0.60 - 9.19)
0.22
2.62 (0.45 - 15.37)
0.29
2.72 (0.17 - 43.18)
0.48
0.05 (0.00 - 0.89)
0.041
ORR (%)
20 (57.1%)
31 (67.4%)
15 (71.4%)
24 (63.2%)
11 (73.3%)
18 (66.7%)
3 (75.0%)
6 (54.5%)
5 (35.7%)
7 (87.5%)
All regions
All regions excl Japan
Europe
North America
Japan
cPP Population
Randomized Treatment Arfolitixorin Leucovorin

Progression-free Survival (cPP)

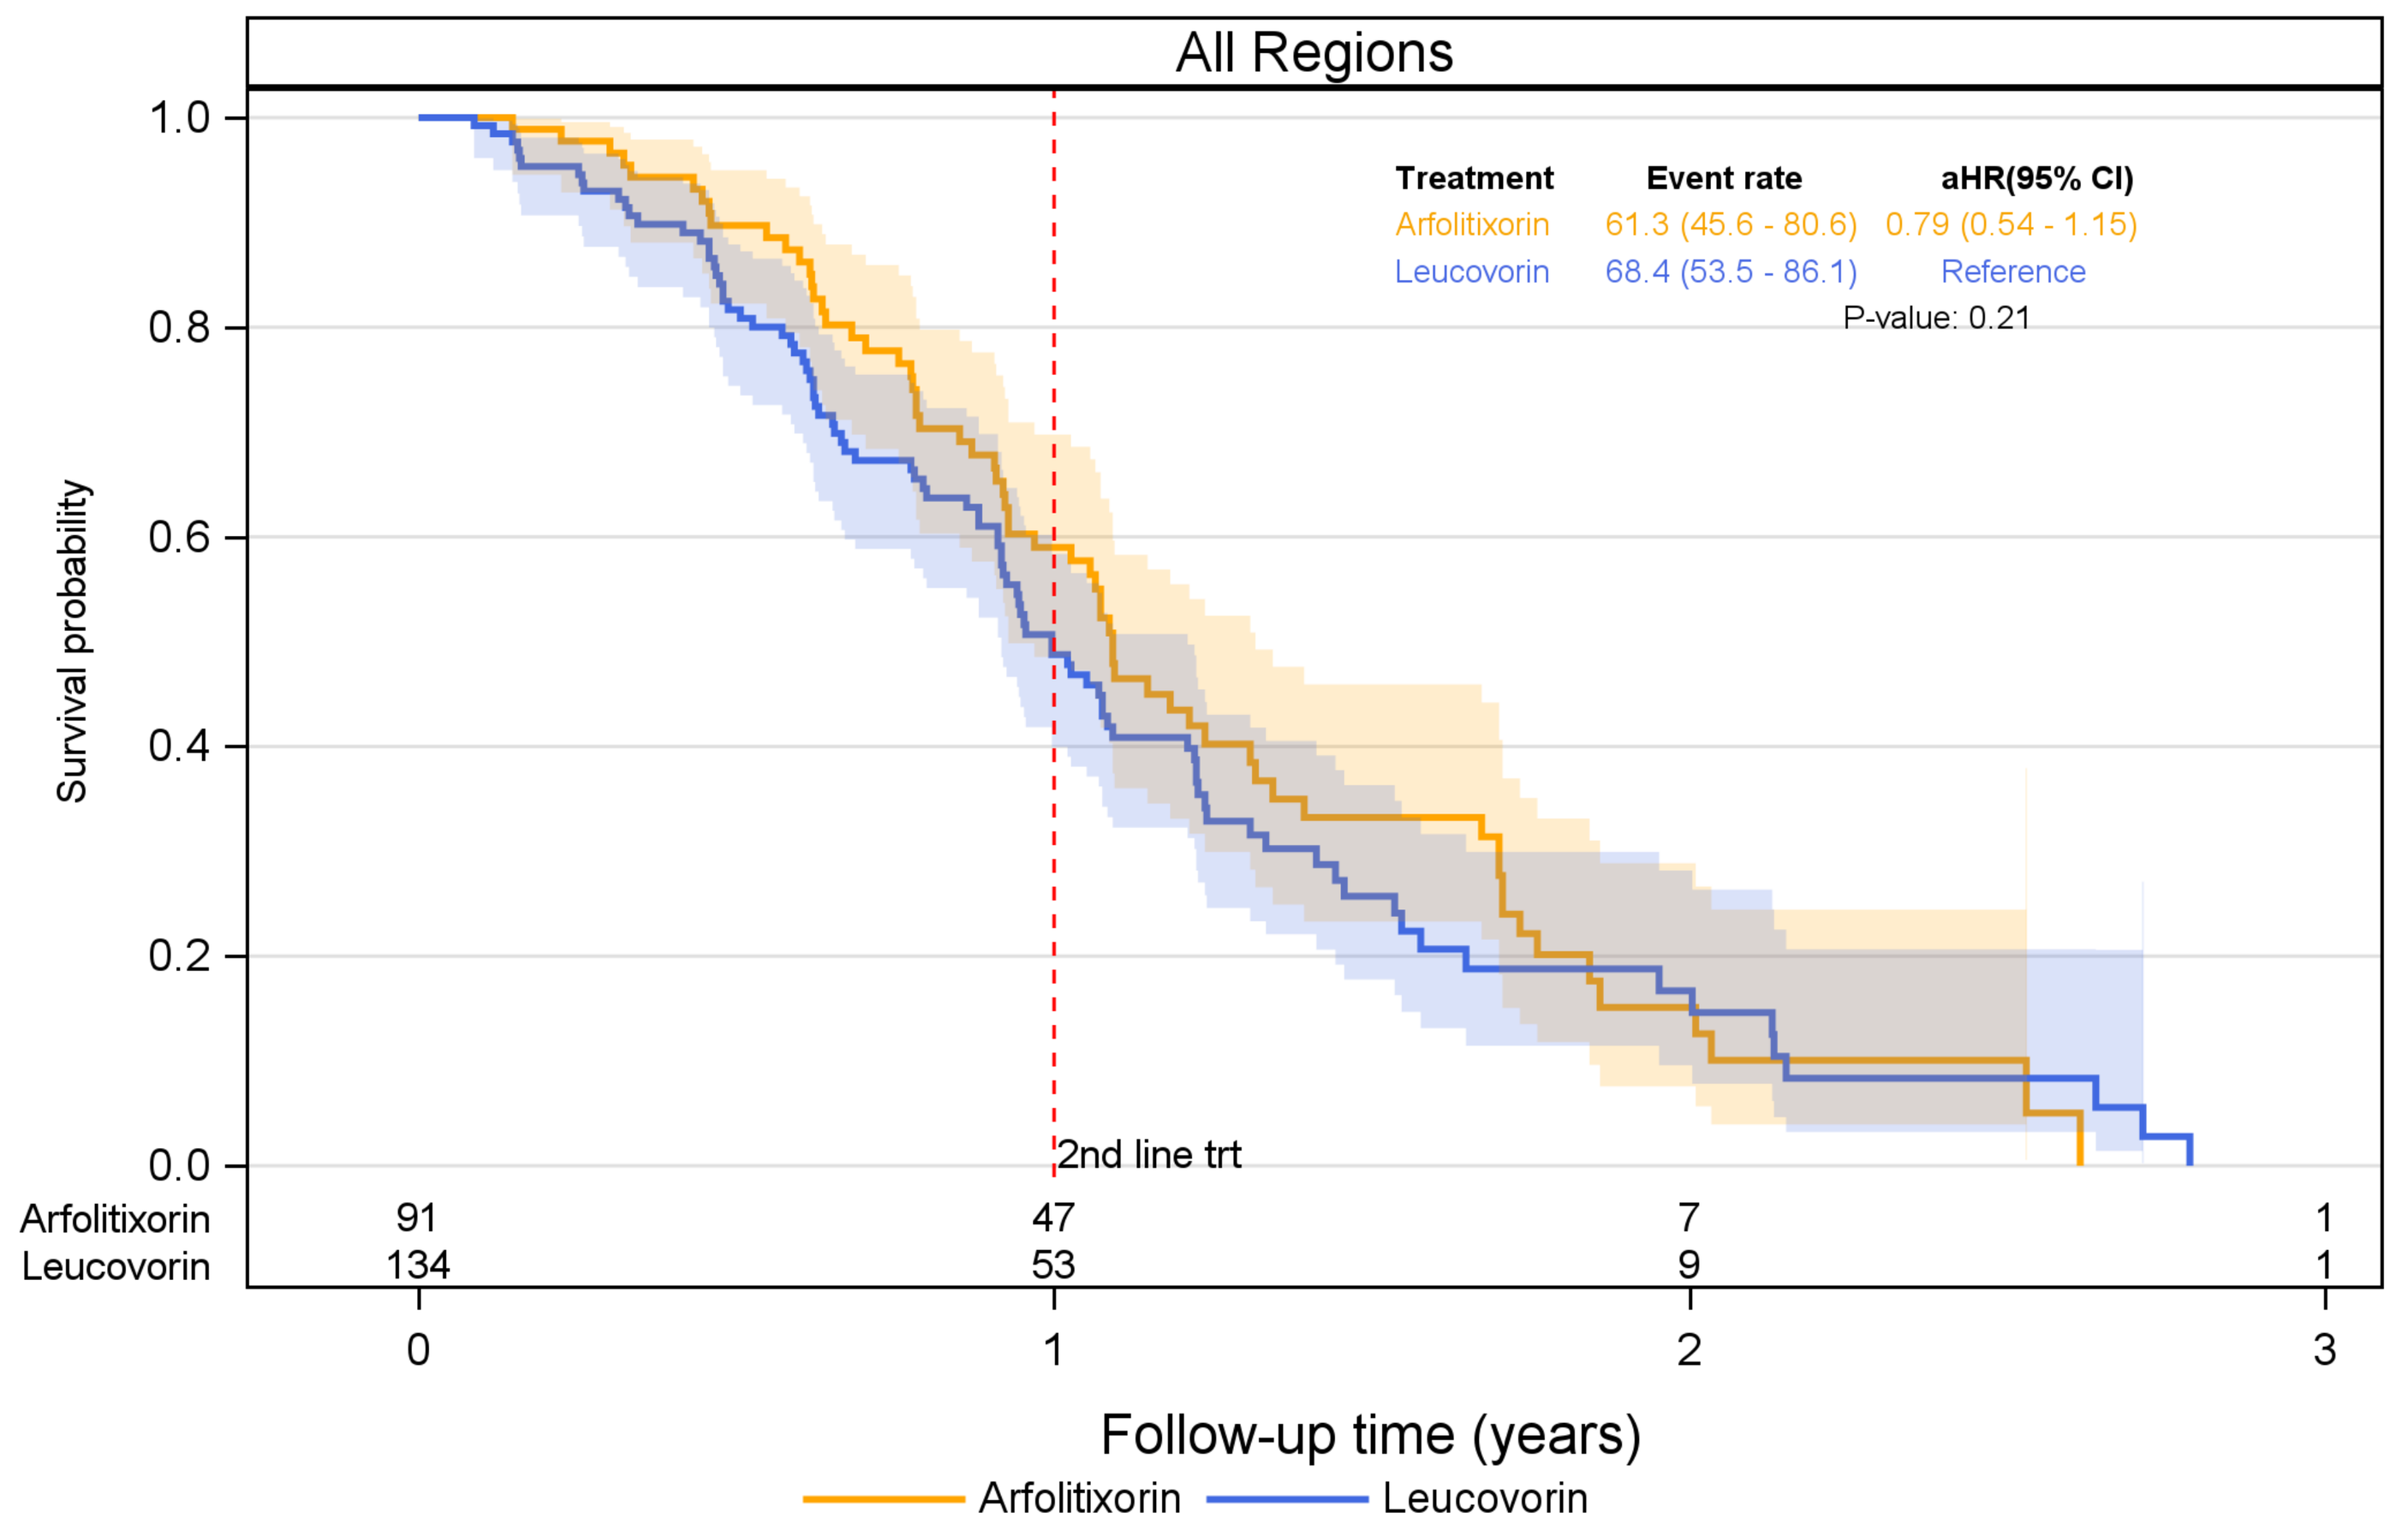
All Regions
Treatment Event rate aHR(95% CI)
Arfolitixorin 61.3 (45.6 - 80.6) 0.79 (0.54 - 1.15)
Leucovorin 68.4 (53.5 - 86.1) Reference
P-value: 0.21
Survival probability
2nd line trt
Arfolitixorin 91 47 7 1
Leucovorin 134 53 9 1
Follow-up time (years)
Arfolitixorin
Leucovorin

Overall Survival (cPP)

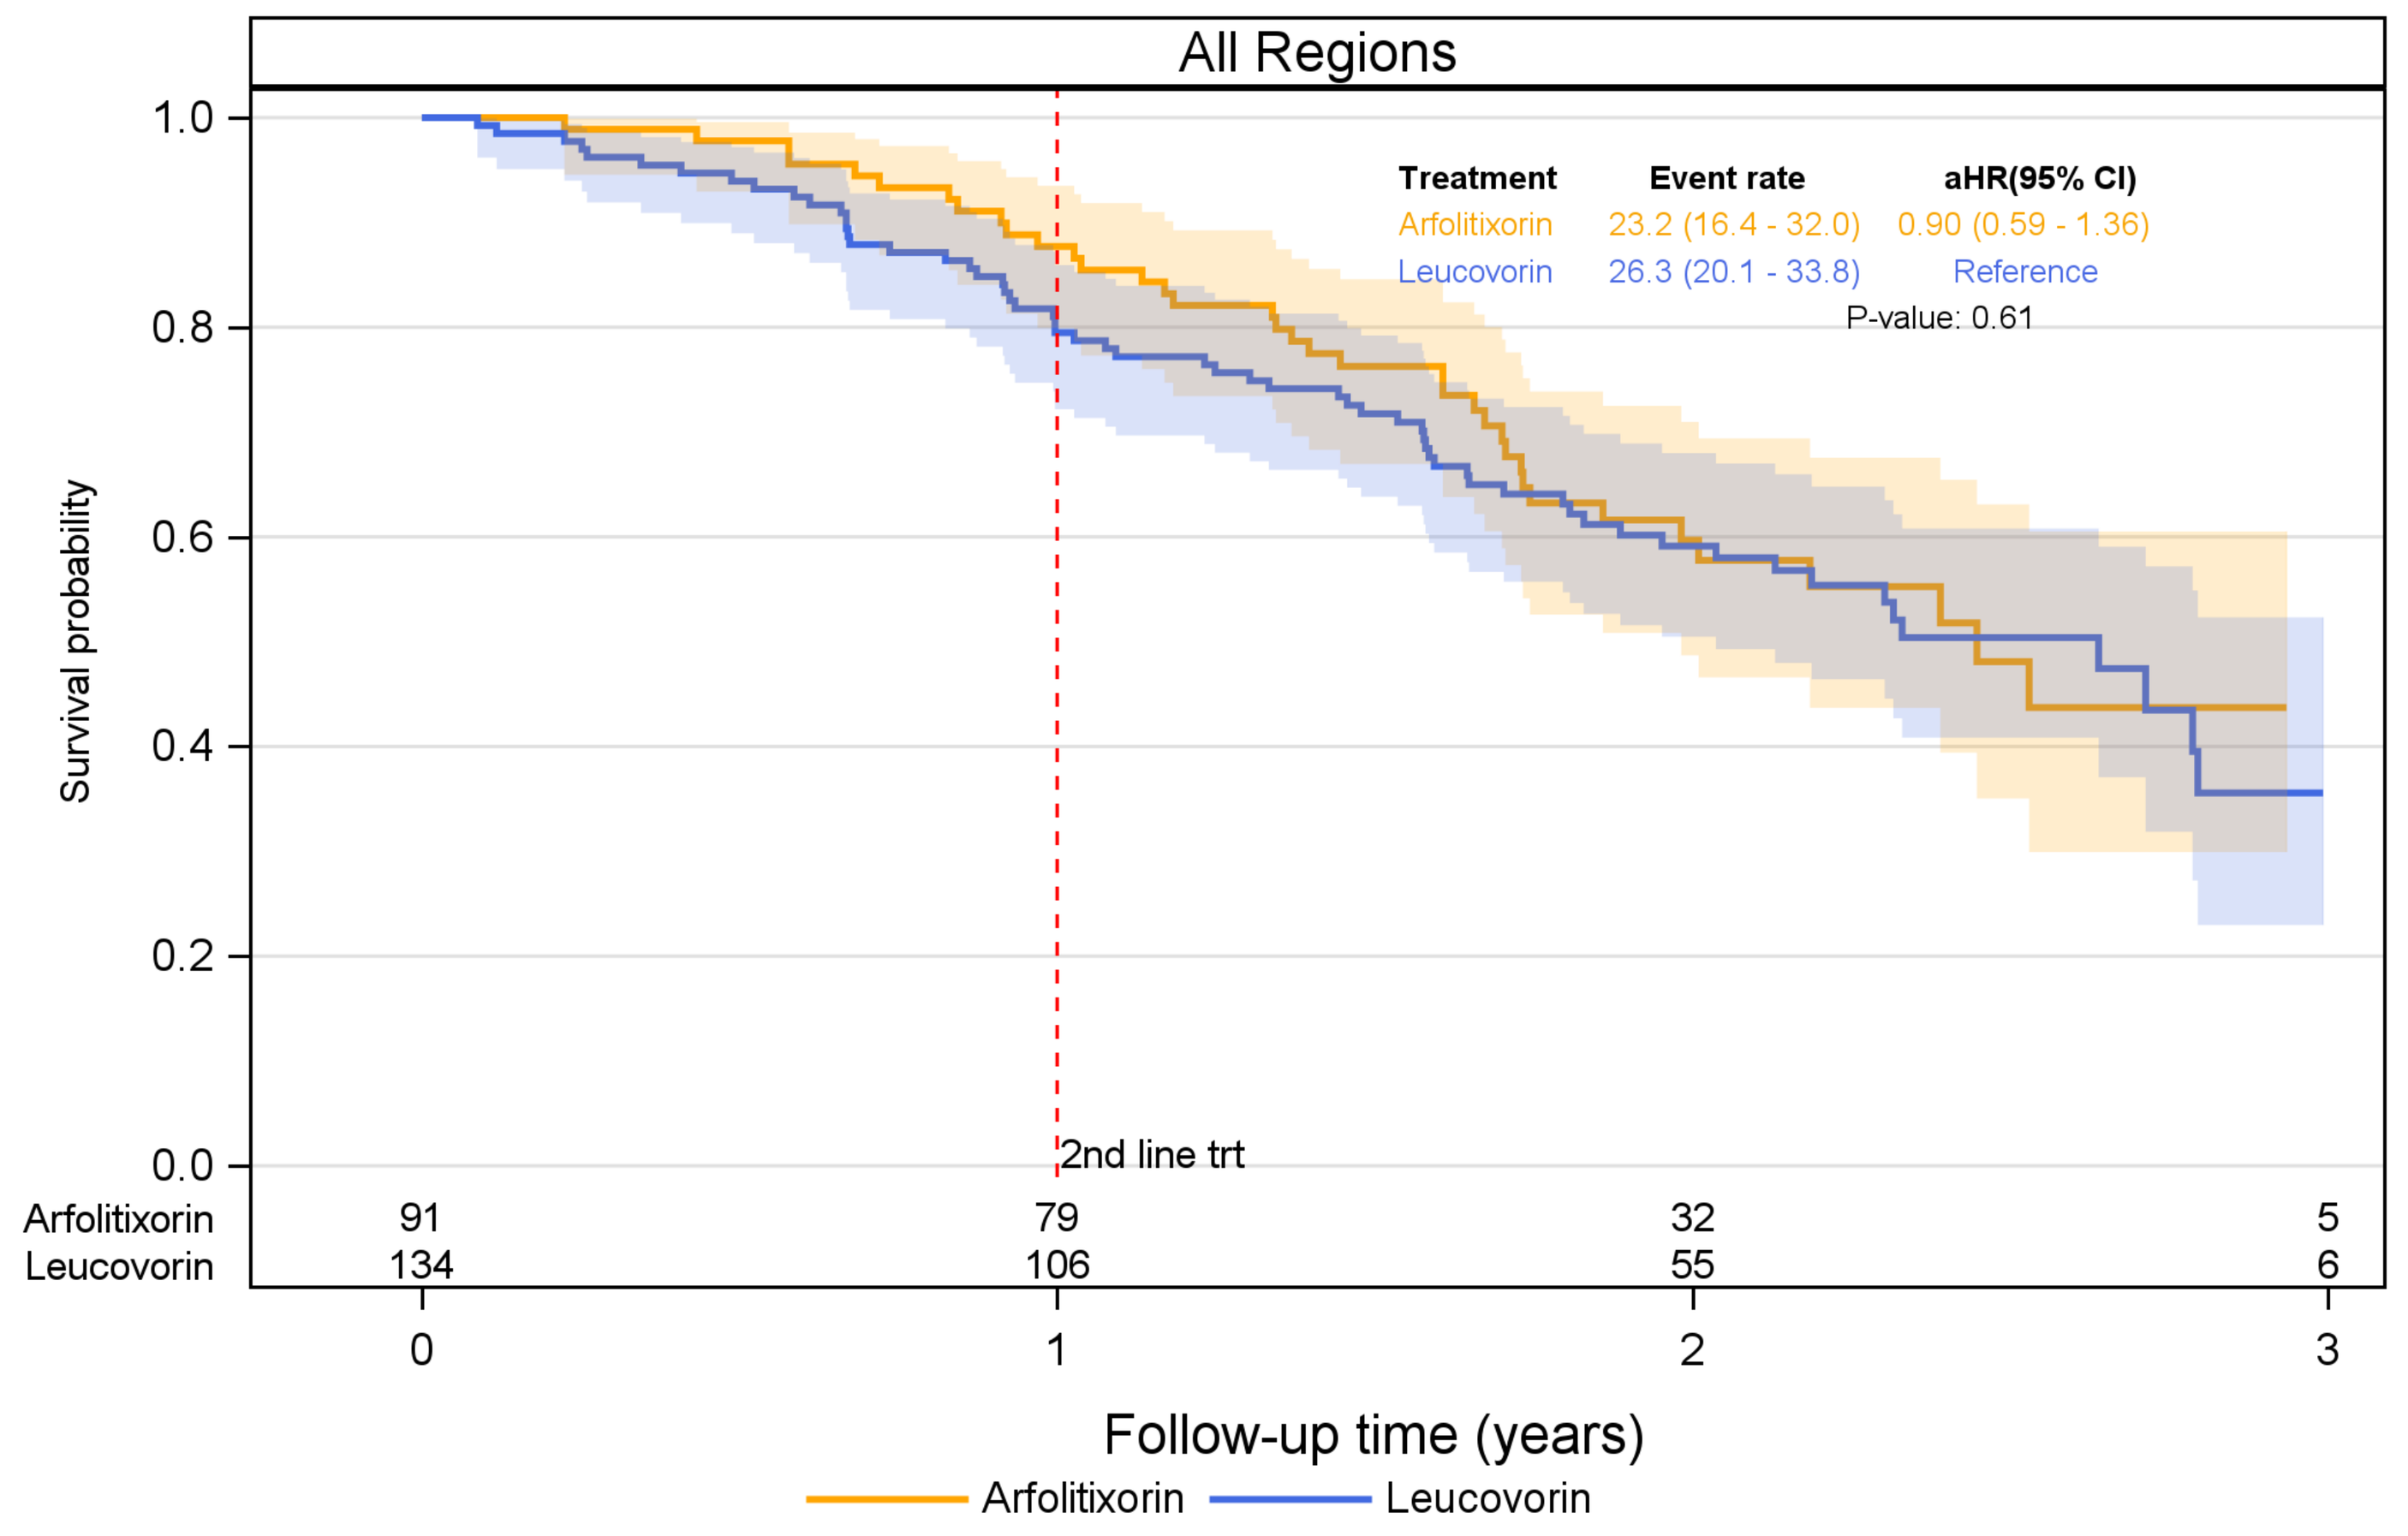
All Regions
Treatment Event rate aHR(95% CI)
Arfolitixorin 23.2 (16.4 - 32.0) 0.90 (0.59 - 1.36)
Leucovorin 26.3 (20.1 - 33.8) Reference
P-value: 0.61
Survival probability
2nd line trt
Arfolitixorin 91 79 32 5
Leucovorin 134 106 55 6
Follow-up time (years)
Arfolitixorin
Leucovorin